Increases in Erythropoiesis-Stimulating Agent Use After Hospitalization of End-Stage Renal Disease Patients

TC Bond, PhD[1]; S Wang, MS[1]; J Rubin, MA[1]; A Yang, MD[2]

[1]DaVita Clinical Research, Minneapolis, MN, USA; [2]Affymax, Inc., Palo Alto, CA, USA

Introduction

- Hemodialysis patients are frequently hospitalized, with a national mean of 1.9 hospitalizations per patient-year in 2009 according to the USRDS 2011 Annual Data Report.[1]
- In 2009, 36% of hospitalizations in hemodialysis patients were followed by another hospitalization within 30 days.[1] Post-hospitalization anemia control has been shown to reduce re-hospitalization rates.[2]
- A combination of the reason for hospitalization and an interruption of normal dialysis treatment leads to lowered hemoglobin levels and increased utilization of erythropoiesis-stimulating agents (ESAs) in the post-hospitalization period.[3]
- Solid *et al*. found that Hb levels take 2 months to recover to pre-hospitalization levels and that optimum ESA usage over this time period has yet to be determined.[4] Brophy *et al*. confirmed that < 20% of dialysis patients receive any ESA dose while hospitalized.[5]

Objective

We have carried out a retrospective analysis to assess changes in Hb levels and ESA utilization in end-stage renal disease patients post-hospitalization.

Methods

- Data from adult (> 18 years old) hemodialysis patients receiving in-center dialysis ≥ 3 times/week between January 1, 2009 and December 31, 2010 were assessed.
- Data analysis was restricted to hospitalizations that occurred ≥ 30 days after a previous hospitalization discharge.
- Mean ESA dose and Mean Hb levels for the 30 days before and 30 days after hospitalization were assessed.
- Patients who experienced a mean rise in ESA dose in the 30 days after hospitalization were assessed to determine whether such changes were:
 - Presumptive - dose increase was observed before the first post-hospitalization Hb test result was known, or
 - Responsive - dose increase occurred on or after the date the first post-hospitalization Hb test result was known.

Results

Table 1. Summary Statistics: Hospitalizations

	Mean ± SD
Patients	138,762
Hospitalizations	289,042
Length of stay (days)	
Mean ± SD	7.76 ± 10.36
Median	5
Time between hospitalizations (days)	
Mean ± SD	74.1 ± 96.7
Median	37

Table 2. Hemoglobin Levels Pre- and Post-Hospitalization

	Number of Hospitalizations	Mean Pre-Hospitalization Hb Level	Mean Post-Hospitalization Hb Level	Mean Change in Hb Level
All	156,353 (100%)	11.48 g/dL	10.88 g/dL	-0.59 g/dL
Hb drop	117,455 (66.7%)	11.87 g/dL	10.55 g/dL	-1.32 g/dL
Hb unchanged	1,149 (0.7%)	11.25 g/dL	11.25 g/dL	–
Hb rise	57,595 (32.7%)	10.67 g/dL	11.56 g/dL	+0.88 g/dL

Table 3. ESA Dose Per-Session in the 30 Days Pre- and Post-Hospitalization

	Number of Hospitalizations	Mean Pre-Hospitalization ESA Dose	Mean Post-Hospitalization ESA Dose	Mean Change in ESA Dose (SD)
All	179,929 (100%)	8,213 U	9,535 U	+ 1,322 U (5,161)
ESA rise	109,420 (60.8%)	6,733 U	10,825 U	+ 4,091 U (3,918)
ESA unchanged	8,740 (4.9%)	10,890 U	10,890 U	–
ESA drop	61,769 (34.3%)	10,456 U	7,061 U	- 3,395 U (3,784)

- Of a total of 289,042 hospitalizations, 181,595 (62.8%) occurred > 30 days after any previous event, and qualified for inclusion (Table 1).
- Analysis of Hb levels was restricted to hospitalizations where Hb test results for the pre- and post-hospitalization period were available (N = 156,353). Of these, 66.7% of events were associated with a drop in Hb (mean of 11.87 g/dL pre-hospitalization to 10.55 g/dL post-hospitalization) (Table 2).
- For hospitalizations where ESA dose data for the pre- and post-hospitalization periods were available (N = 179,929), 60.8% of events were associated with a mean rise in ESA dose. For these 109,420 events, the mean ESA dose was 6,733 U/session pre-hospitalization, and 10,825 U/session post-hospitalization, a mean rise of 4,091 U/session (SD = 3,918) (Table 3 and Figure 1).
- Of the 109,420 hospitalizations with mean per-session increases in ESA dose in the post-hospitalization period, 76,242 (69.7%) of these rises were presumptive (ESA dose change made before first post-hospitalization Hb test result obtained) and 33,178 (30.3%) were responsive (dose change occurred after first post-hospitalization Hb test result was obtained).
- Among the 76.242 hospitalizations that were associated with a responsive rise in ESA dose:
 - 23,946 (72.2%) experienced a drop in ESA before first test
 - 9,014 (27.2%) had no change in ESA before first test
 - 218 (0.7%) did not have an ESA dose before their first test

Figure 1. Changes in ESA Dose Post-Hospitalization

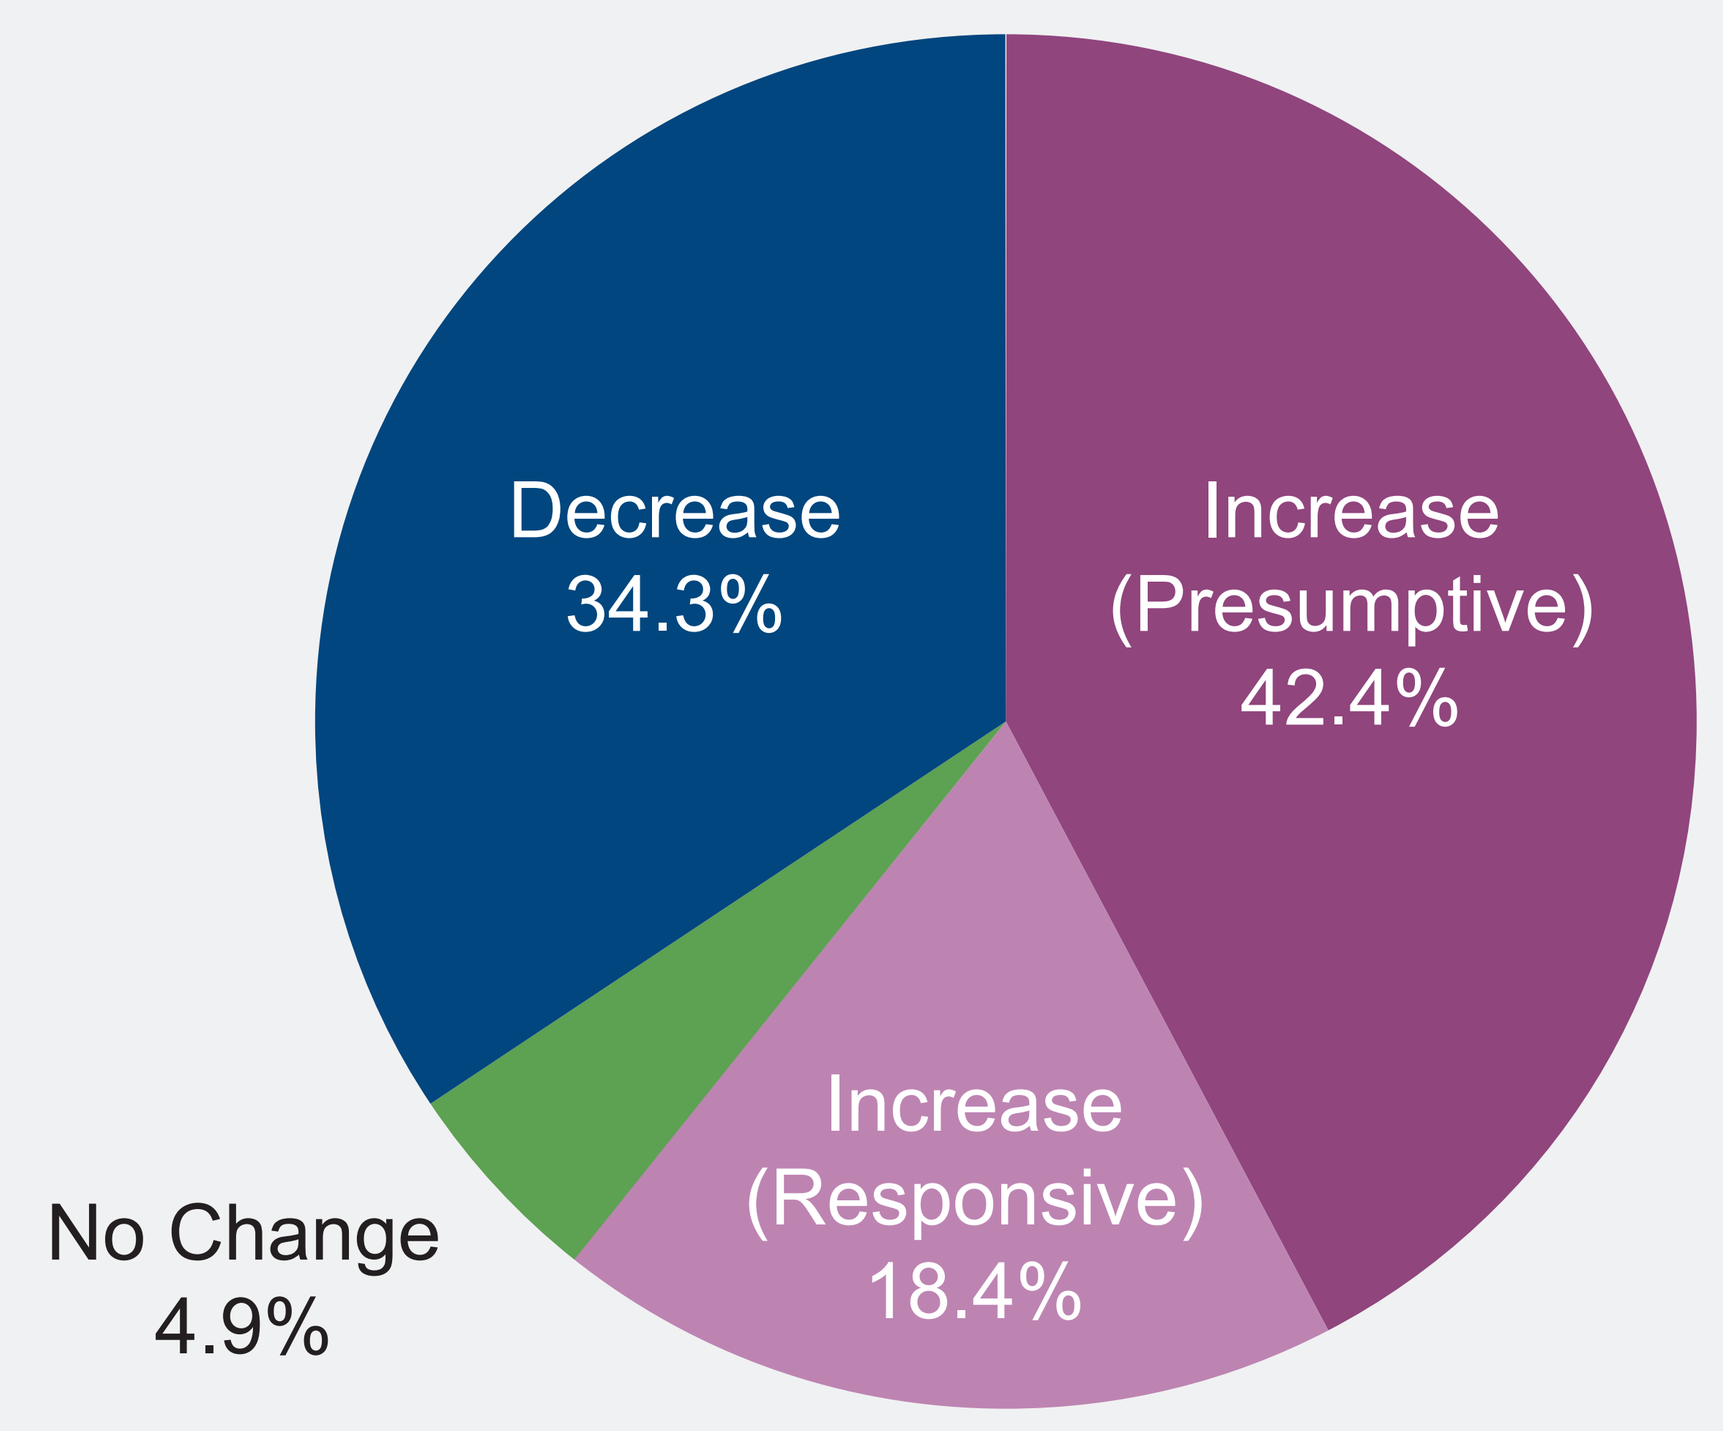
Decrease 34.3%
Increase (Presumptive) 42.4%
Increase (Responsive) 18.4%
No Change 4.9%

Conclusions

- Hospitalizations frequently result in a drop in Hb levels and an increase in ESA dose in the post-hospitalization period.
- Two thirds (66.7%) of hospitalization events were associated with a drop in Hb levels in the 30 days following hospitalization compared to the 30 days before, with a mean change in Hb level of -1.32 g/dL.
- Approximately 60% of hospitalization events were associated with a rise in ESA dose in the 30 days post-hospitalization compared to the 30 days before, with a mean dose change of +4,091 U/session.
- The majority (69.7%) of post-hospitalization increases in ESA dose occur before the result of the first post-hospitalization Hb test is known.
- Strategies for better management of anemia during hospitalization and in the post-hospitalization period should be assessed.

References

1. U.S. Renal Data System. USRDS 2011 Annual Data Report: Atlas of Chronic Kidney Disease and End-Stage Renal Disease in the United States. National Institutes of Health, National Institute of Diabetes and Digestive and Kidney Diseases, Bethesda, MD, 2011.
2. Chan, KE, Lazarus, JM, Wingard, RL, Hakim, RM. Association between repeat hospitalization and early intervention in dialysis patients following hospital discharge. *Kidney Int.* 2009;76:331–341.
3. Speigel D, Gitlin M, Mayne TJ. Factors affecting anemia management in hemodialysis patients: A single-center experience. Hemodialysis Int. 2008;12:336-341.
4. Solid CA, Foley RN, Gilbertson DT, Collins AJ. Perihospitalization hemoglobin-epoetin associations in U.S. hemodialysis patients, 1998 to 2003. *Hemodial Int.* 2007;11(4):442–7.
5. Brophy DF, Daniel G, Gitlin M, Mayne TJ. Characterizing hospitalizations of end-stage renal disease patients on dialysis and inpatient utilization of Erythropoiesis-Stimulating Agent therapy. *Ann Pharmacother* 2010;44(1):43–9.

Acknowledgements

Our sincere appreciation is extended to the teammates in more than 1,800 DaVita clinics who work every day to take care of patients but also to ensure the extensive data collection on which our work is based. We thank DaVita Clinical Research® (DCR®), and specifically acknowledge Abigail Hunt, PhD of DCR for editorial contributions in preparing this poster. DCR is committed to advancing the knowledge and practice of kidney care.

This analysis was funded by Affymax Inc. and Takeda Pharmaceutical Company Limited.

*Correspondence: t.christopher.bond@davita.com

Poster available at www.davitaclinicalresearch.com/directory.asp

The 16th International Congress on Renal Nutrition and Metabolism, Honolulu, Hawaii; June 26–30, 2012

©2012 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted or distributed without the permission of DaVita Inc.